Preliminary Safety and Pharmacokinetics of the MET-TKI DO-2 in Patients with Advanced Solid Tumors Harboring MET Aberrations: A Phase I Study

ENA 2024 EORTC NCI AACR 36th Symposium

PROTOCOL DO2.22.01
NCT05752552

Poster 143

B. Sikkema[1], B. Dekeyser[2], P. de Bruijn[1], F. Zammit[3], R. Galot[3], J.P. Machiels[3], B. Delafontaine[4], S. Rottey[4], A. Swalduz[5], F. Ghiringhelli[6], C. van Herpen[7], I. Desar[7], A. Cortot[8], F. Wastelin[9], T. Perera[9], J. Verweij[9], R. Mathijssen[1], D. Robbrecht[1], H. Prenen[2]

[1]Erasmus MC Cancer Institute, Dept. of Medical Oncology, Rotterdam, The Netherlands; [2]University Hospital Antwerp, Dept. of Medical Oncology, Edegem, Belgium; [3]Institut Roi Albert II Cancer Center, Cliniques Universitaires Saint-Luc UCLouvain, Brussels, Belgium; [4]Ghent University Hospital, Dept. of Medical Oncology, Ghent, Belgium; Centre Leon Berard, Dept. of Medical Oncology, Lyon, France; [6]Centre Georges Francois Leclerc, Department of Medical Oncology, Dijon, France; [7]Radboud University Medical Center, Dept. of Medical Oncology, Nijmegen, The Netherlands; [8]CHU Lille, Institut Cœur Poumon, Lille, France; [9]DeuterOncology SA, Liege, Belgium.

Introduction

- DO-2 is a highly selective type 1B MET TKI re-engineered to block a metabolic liability using selective deuterium replacement.
- Activating mutations of MET kinase including the exon14 skipping mutation result in oncogenic signalling via overexpression of wtMET receptor on tumour cells.
- The degree of inhibition of MET signalling on tumour cells will be matched by 'on target' inhibition of physiological MET signalling.
- DO-2 has 'fast on' and 'fast off' binding kinetics on MET (see poster 407), that results in target inhibition being directly correlated with plasma exposure.
- Preclinical data indicate that maintaining >95% MET target inhibition for 8-10 hours is required and sufficient to drive complete regression of MET driven xenograft models.
- The minimum effective dose in mice is <0.3mg/kg and efficacy correlated with achieving the required time over threshold (ToT) and not AUC_{0-24} or C_{max}
- DO-2 clinical exposure above 160 ng/mL for 8-10hrs (ToT) predicted to drive clinical efficacy.
- Here, we report promising preliminary findings from a first-in-human phase I trial (NCT05752552) of DO-2 in patients with advanced solid tumors harboring MET aberrations, aiming to determine pharmacokinetics, safety and the recommended phase 2 dose (RP2D).

Pharmacokinetics of DO-2

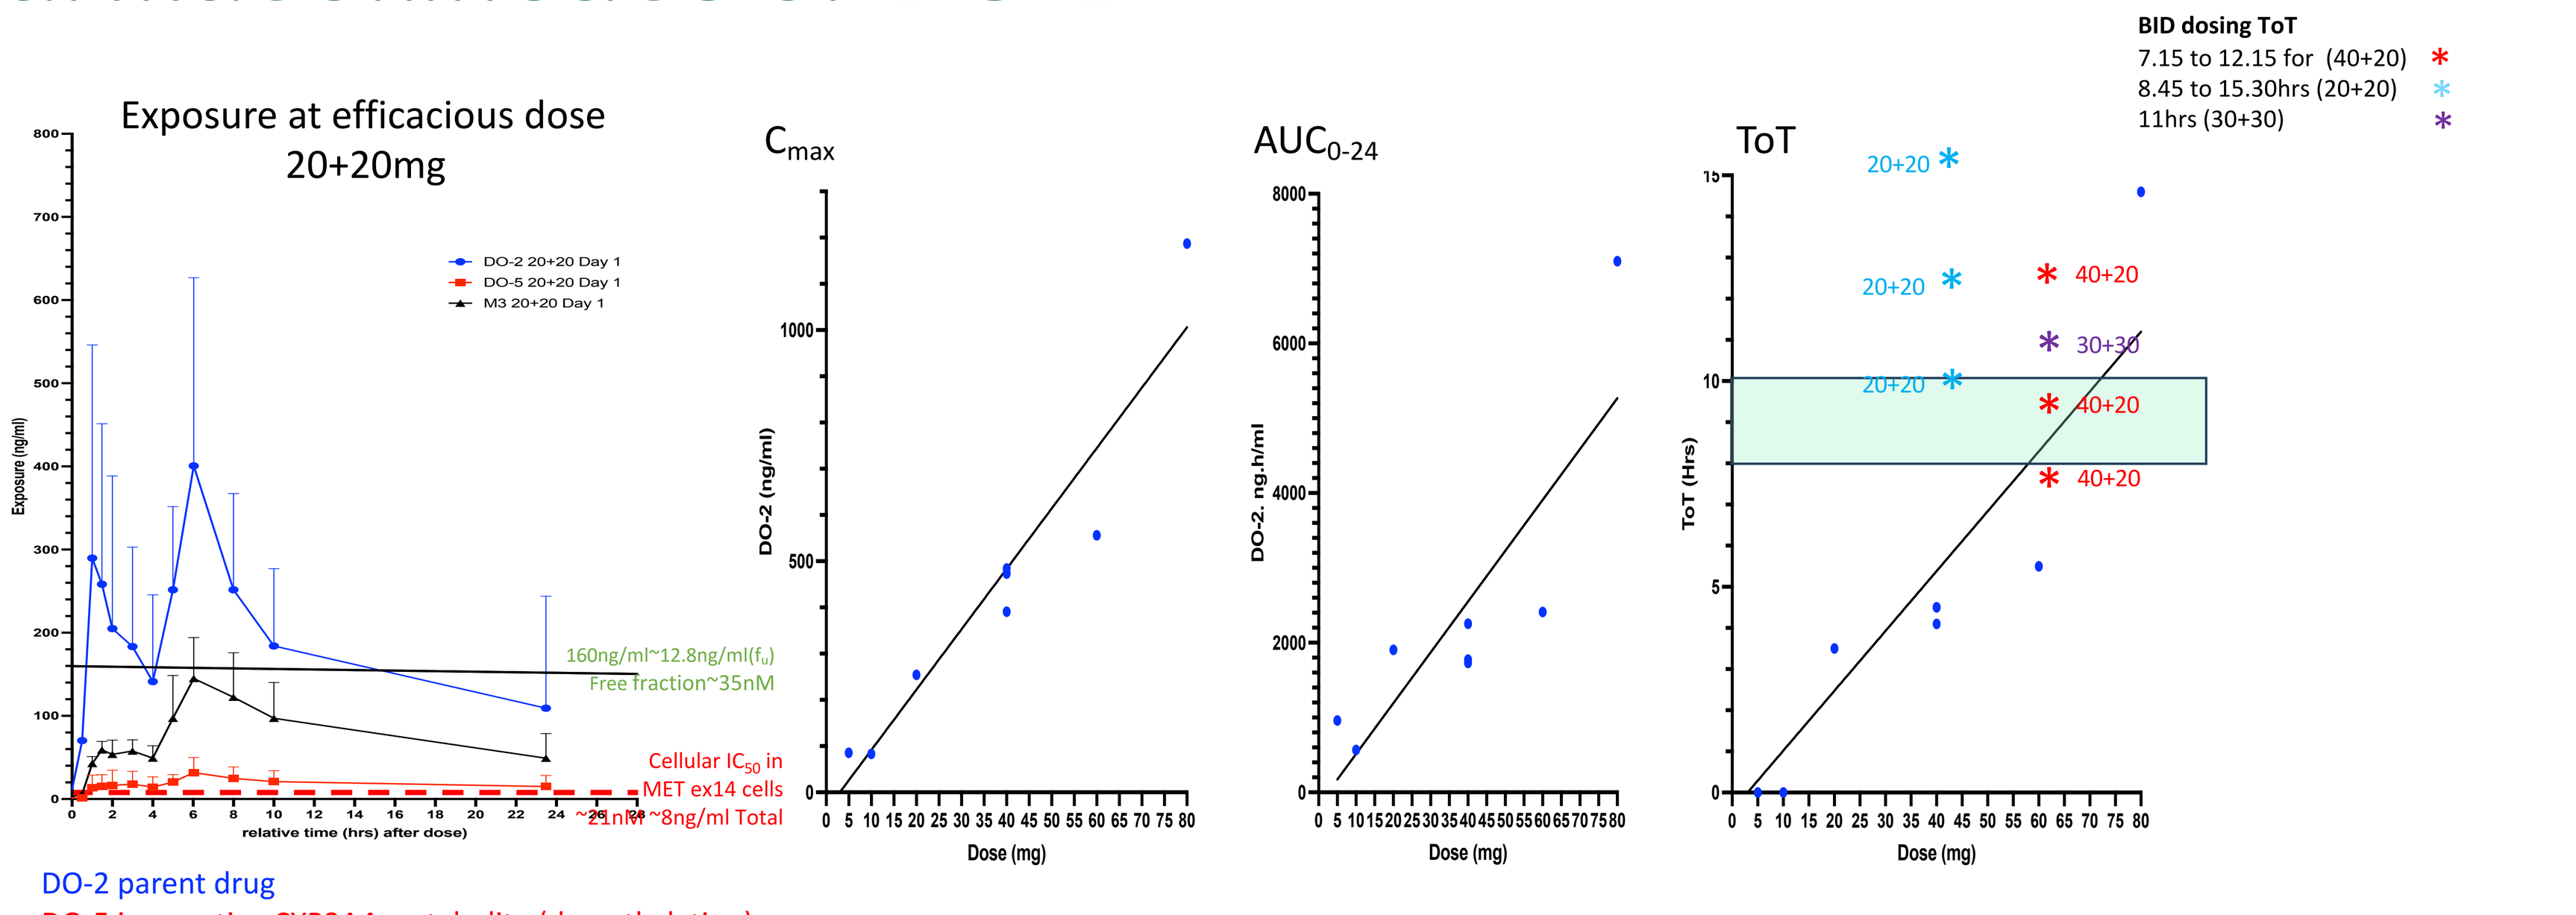
Exposure at efficacious dose 20+20mg
DO-2 20+20 Day 1
DO-5 20+20 Day 1
M3 20+20 Day 1
Exposure (ng/ml)
relative time (hrs) after dose)
160ng/ml~12.8ng/ml(fu)
Free fraction~35nM
Cellular IC50 in MET ex14 cells
~21nM ~8ng/ml Total
Cmax
DO-2 (ng/ml)
Dose (mg)
AUC0-24
DO-2. ng.h/ml
ToT
ToT (Hrs)
BID dosing ToT
7.15 to 12.15 for (40+20)
8.45 to 15.30hrs (20+20)
11hrs (30+30)
20+20
40+20
30+30

DO-2 parent drug
DO-5 is an active CYP3A4 metabolite (demethylation)
M3 is an inactive Aldehyde Oxidase metabolite (Oxidation) that can be further metabolized to form a further metabolite (M5)

- Dose dependent increase in C_{max} and AUC with QD dosing
- ToT exceeded with BID dosing without excessive C_{max}

Analyte	DO-2							
Dose (mg)	5	10	20	40	40	40	60	80
T_{max} (h)	1.5	4	1.43	2.0	1.5	1.6	1.4	2.1
C_{max} (ng/mL)	85.6	82.9	254	473	391	484	556	1187
AUC_{0-24} (ng.h/mL)	959	566	1905	2253	1730	1774	2412	7099
ToT hrs over 160ng/ml	0	0	4	4.5	4.1	4.5	5.5	14.6

Dose escalation design

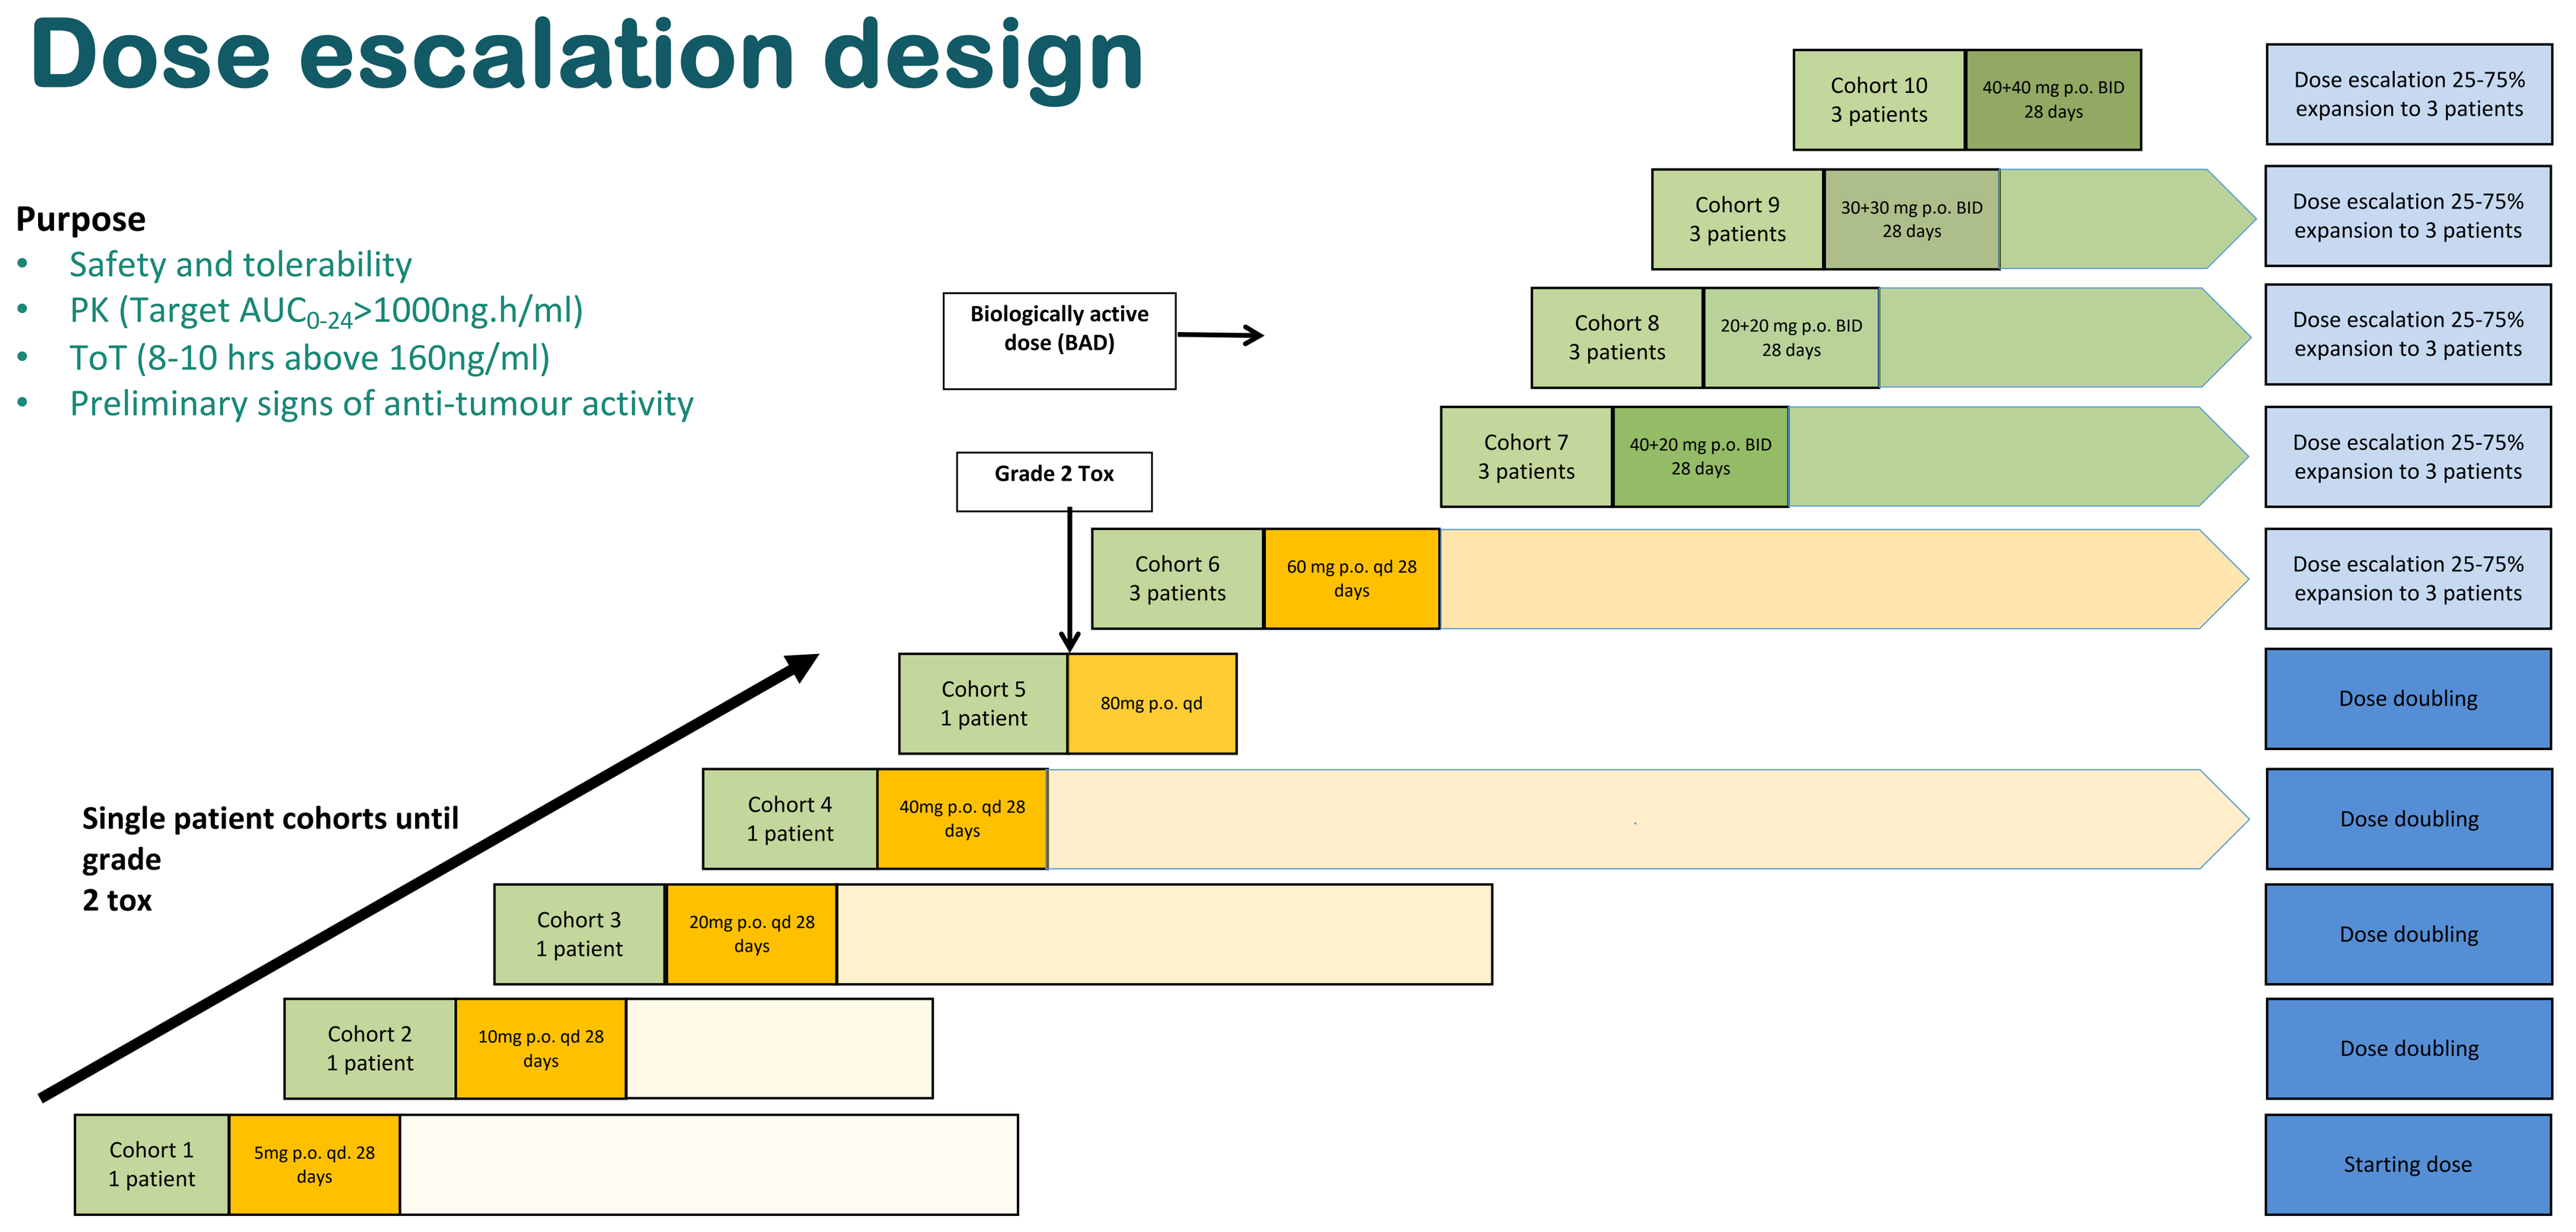
Purpose
Safety and tolerability
PK (Target AUC0-24>1000ng.h/ml)
ToT (8-10 hrs above 160ng/ml)
Preliminary signs of anti-tumour activity
Biologically active dose (BAD)
Grade 2 Tox
Single patient cohorts until grade 2 tox
Cohort 10 3 patients 40+40 mg p.o. BID 28 days
Cohort 9 3 patients 30+30 mg p.o. BID 28 days
Cohort 8 3 patients 20+20 mg p.o. BID 28 days
Cohort 7 3 patients 40+20 mg p.o. BID 28 days
Cohort 6 3 patients 60 mg p.o. qd 28 days
Cohort 5 1 patient 80mg p.o. qd
Cohort 4 1 patient 40mg p.o. qd 28 days
Cohort 3 1 patient 20mg p.o. qd 28 days
Cohort 2 1 patient 10mg p.o. qd 28 days
Cohort 1 1 patient 5mg p.o. qd. 28 days
Dose escalation 25-75% expansion to 3 patients
Dose doubling
Starting dose

Preliminary clinical activity

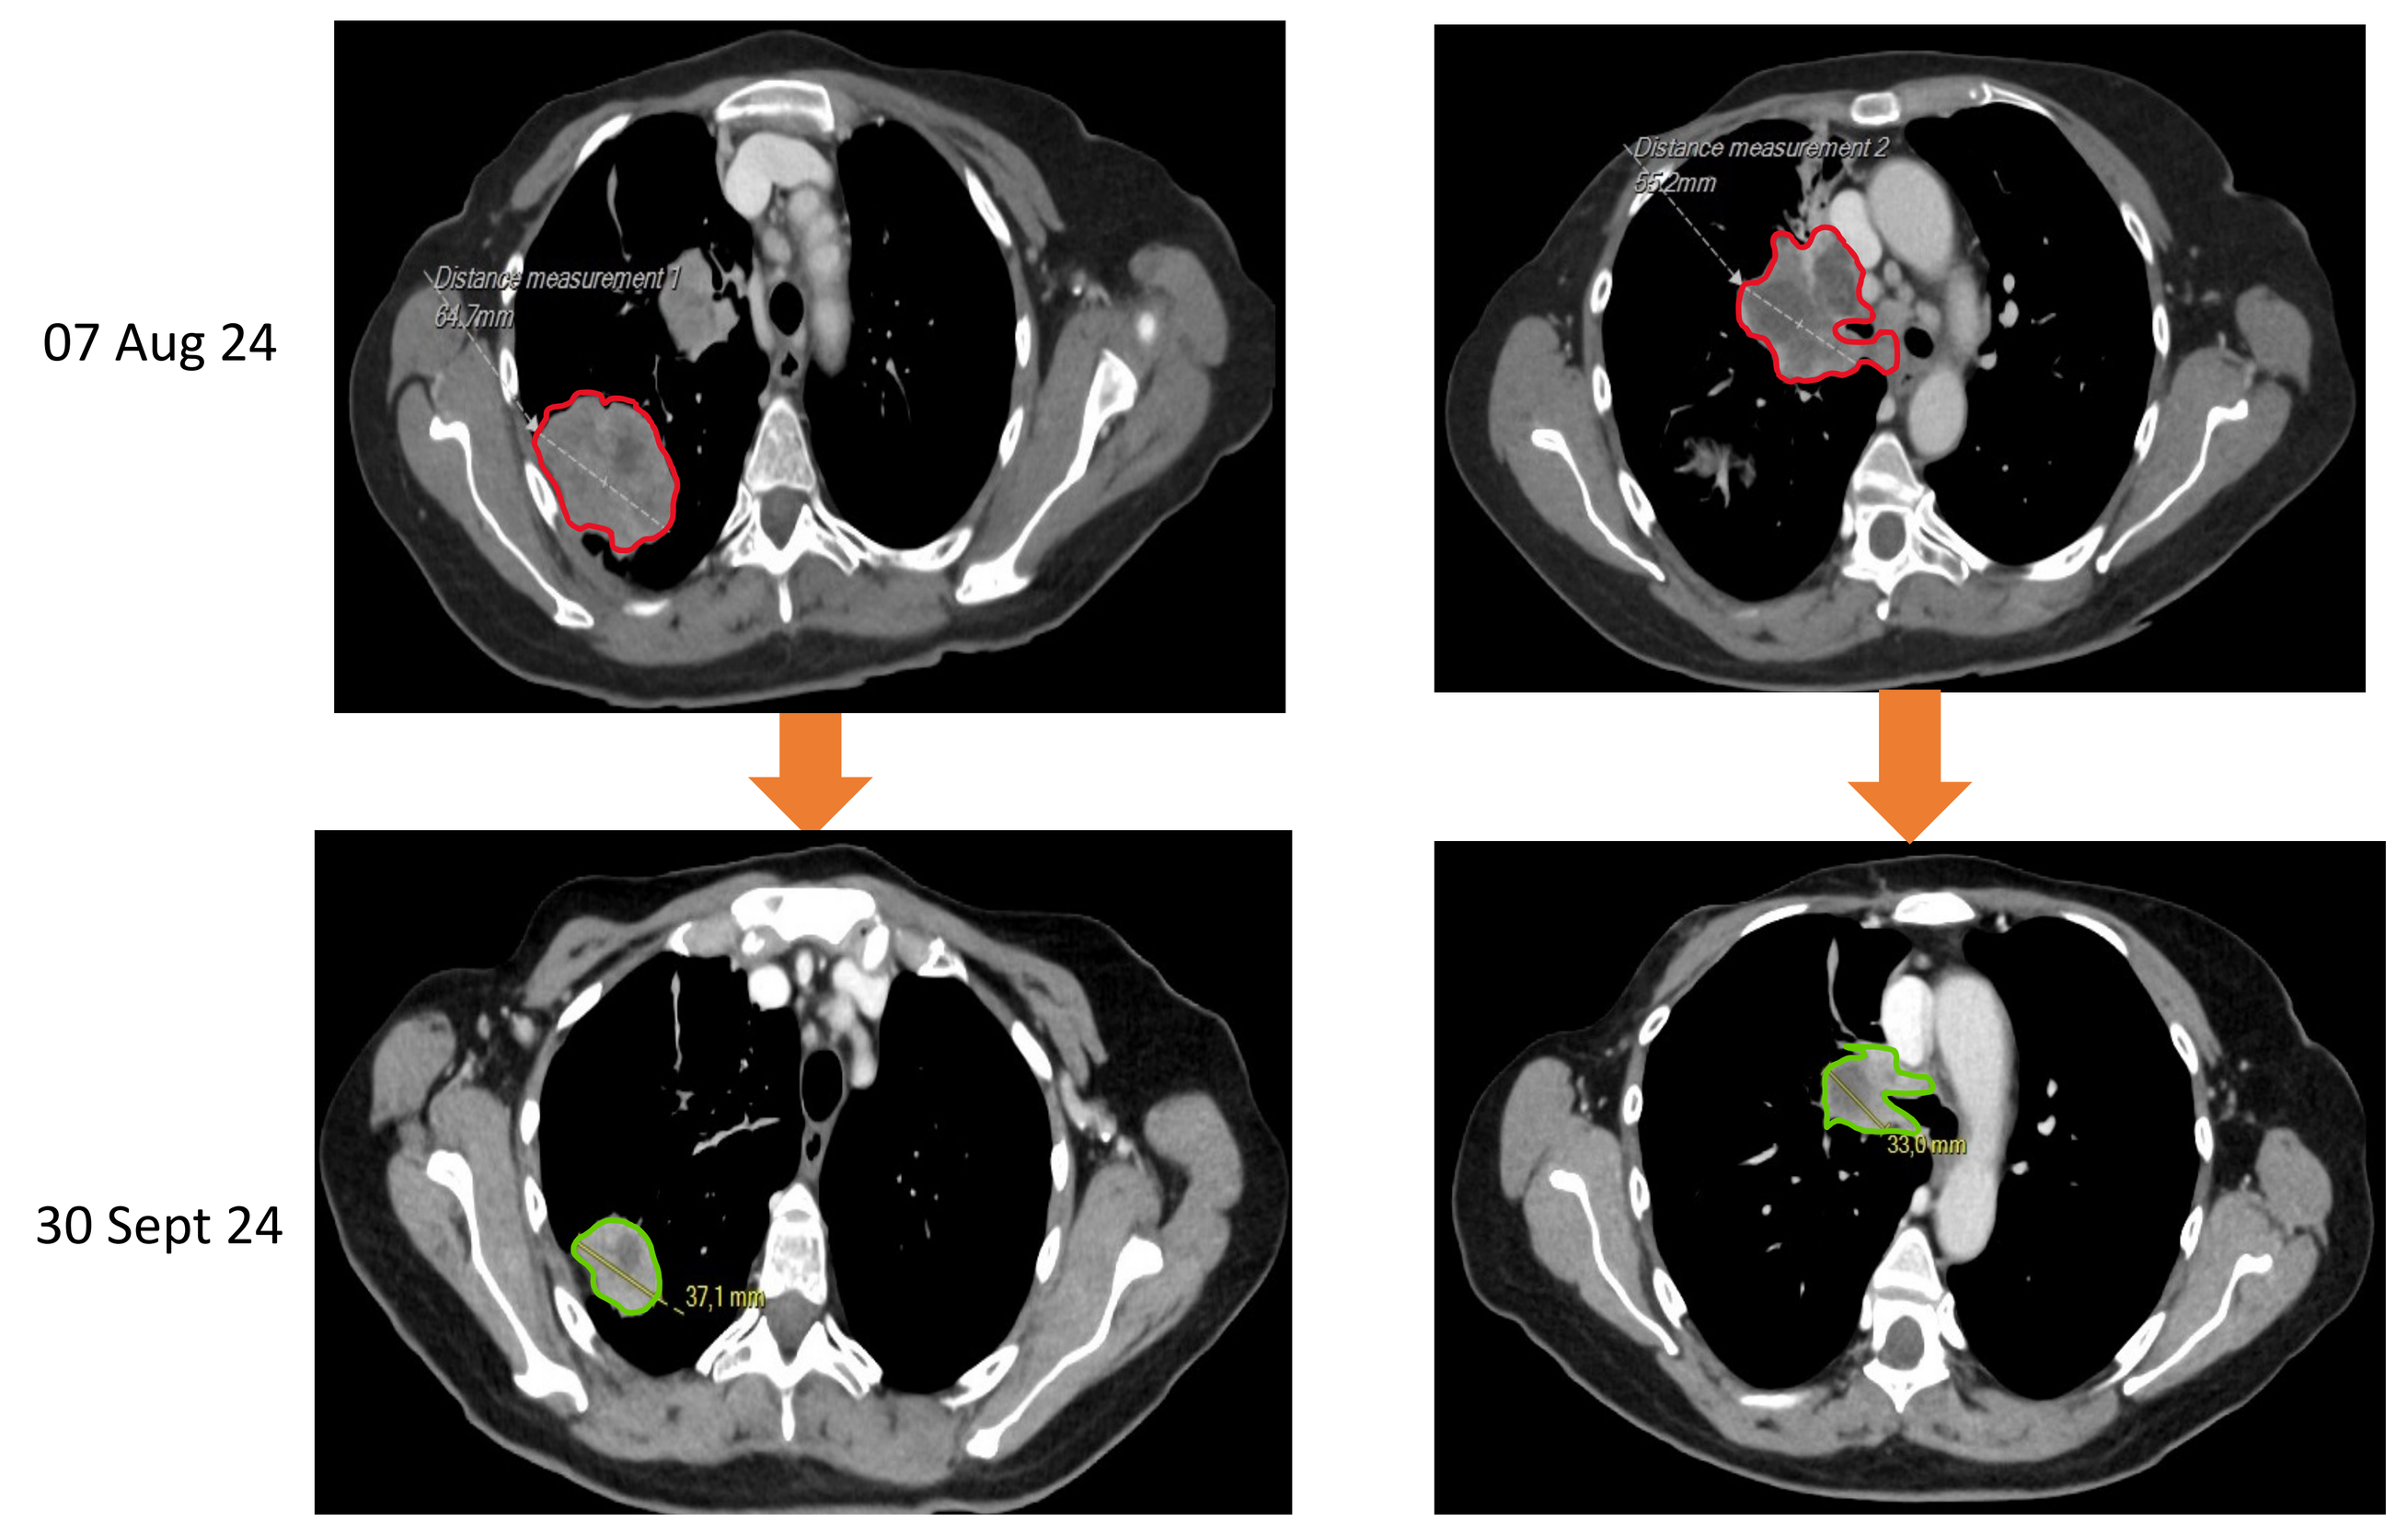
07 Aug 24
30 Sept 24
Distance measurement 1
64.7mm
Distance measurement 2
55.2mm
37,1 mm
33,0 mm

RECIST 1.1
Partial Response (↓39%)

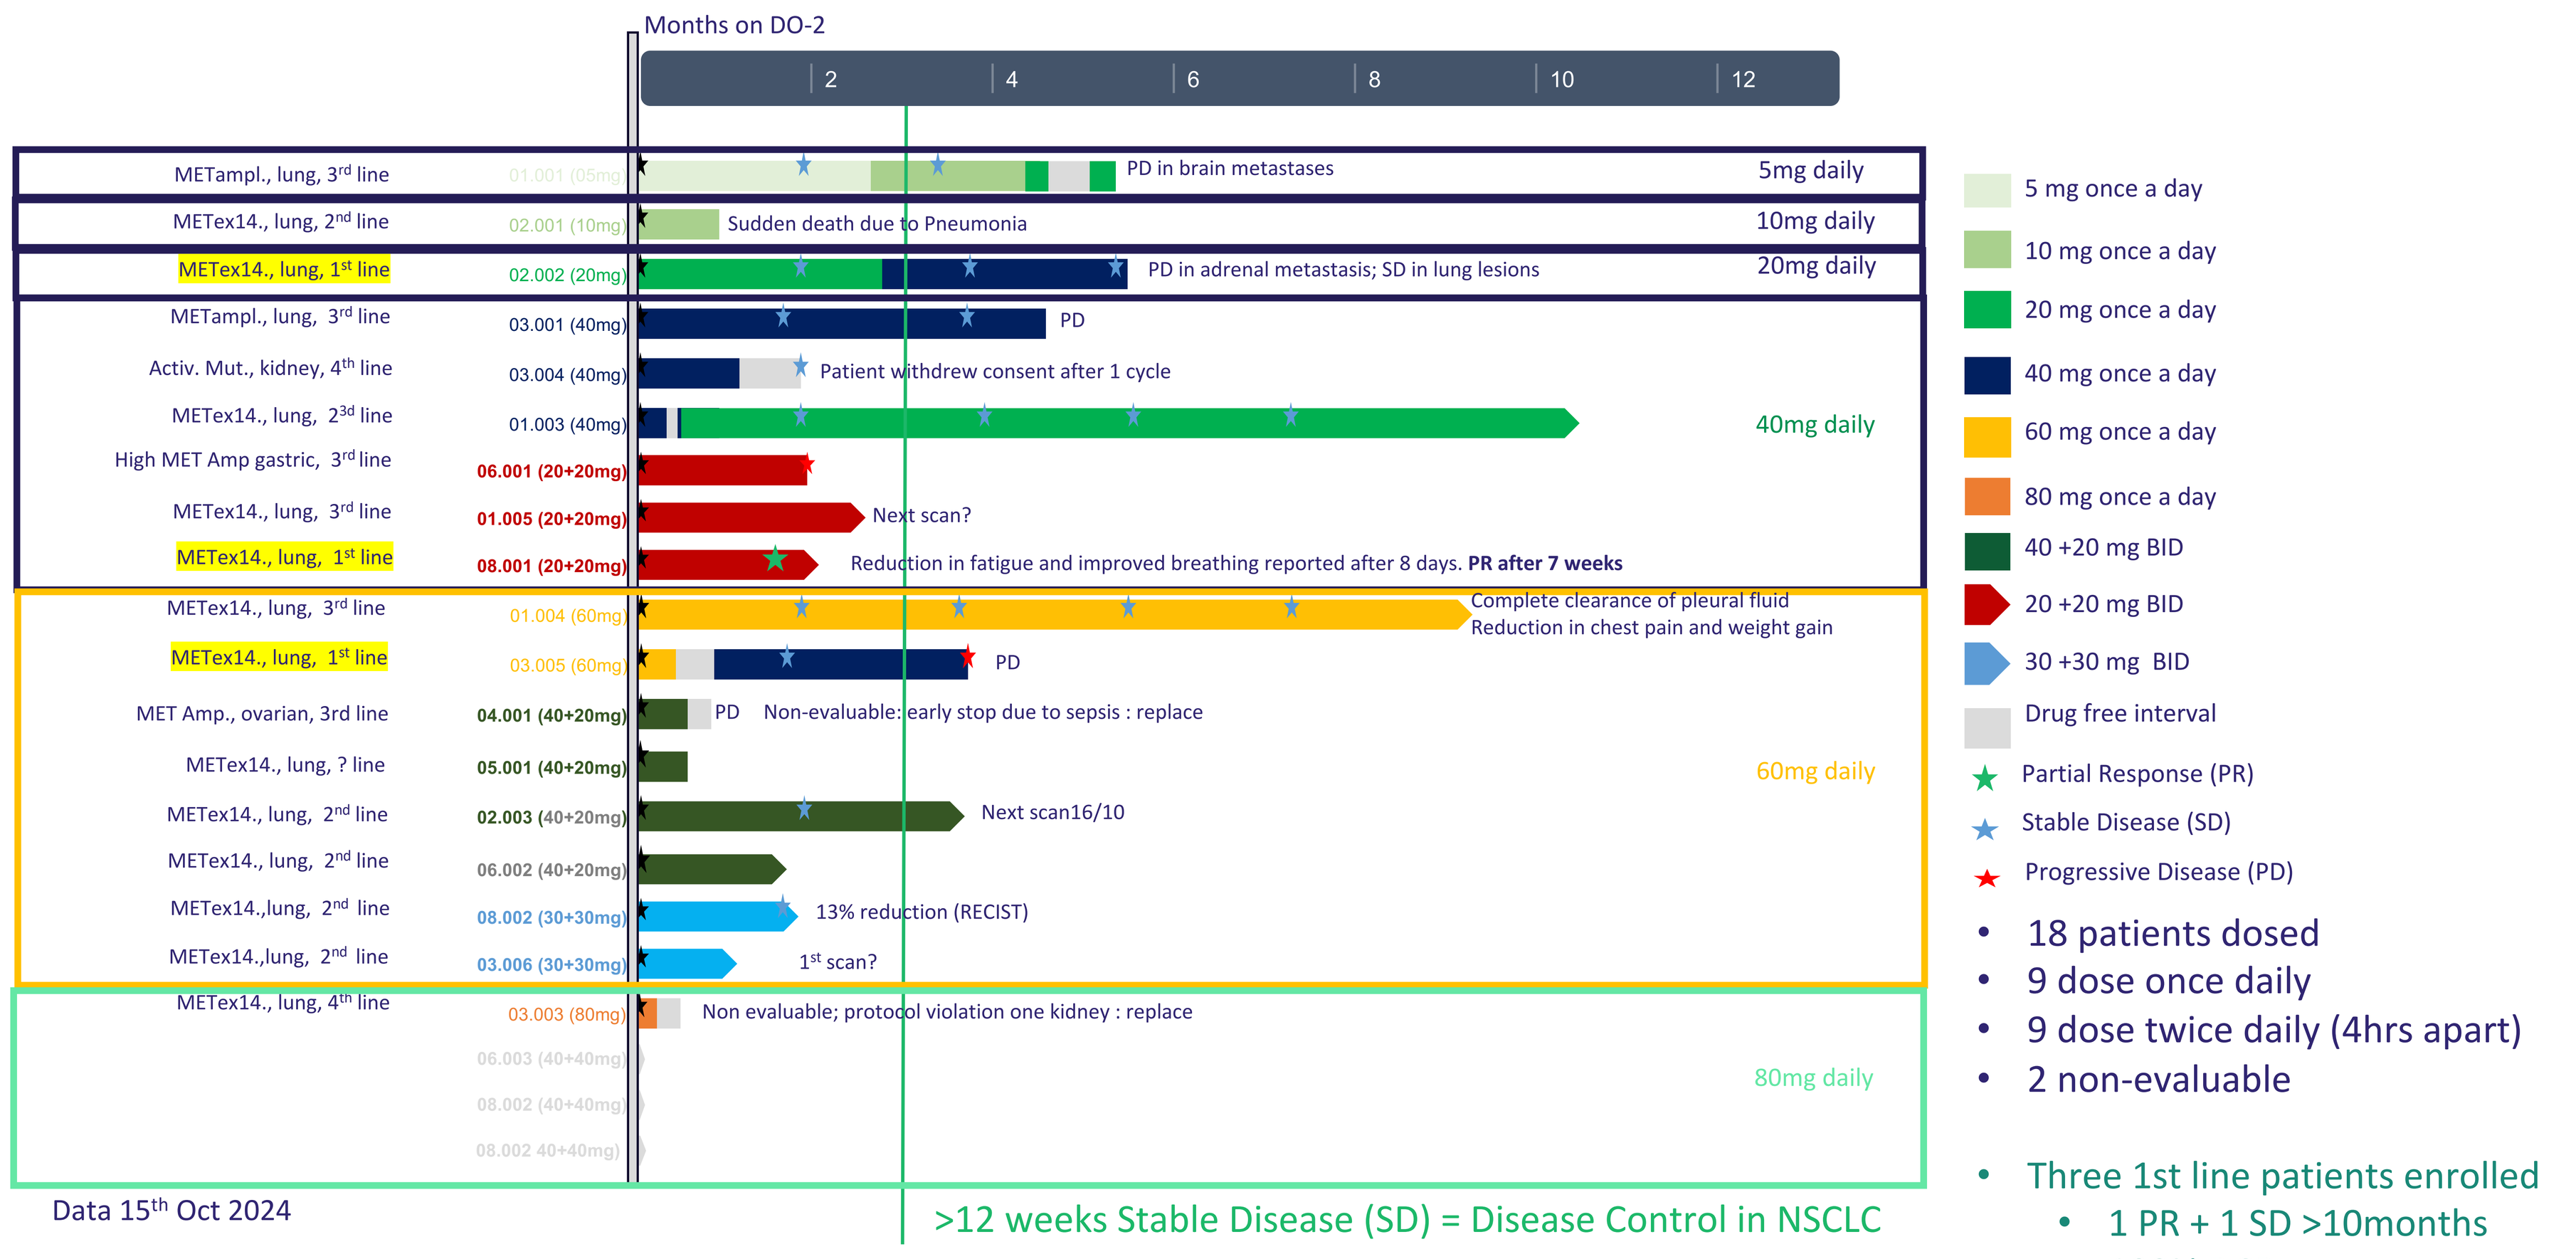
Months on DO-2
2
4
6
8
10
12
METampl., lung, 3rd line 01.001 (05mg) PD in brain metastases 5mg daily
METex14., lung, 2nd line 02.001 (10mg) Sudden death due to Pneumonia 10mg daily
METex14., lung, 1st line 02.002 (20mg) PD in adrenal metastasis; SD in lung lesions 20mg daily
METampl., lung, 3rd line 03.001 (40mg) PD
Activ. Mut., kidney, 4th line 03.004 (40mg) Patient withdrew consent after 1 cycle
METex14., lung, 23d line 01.003 (40mg) 40mg daily
High MET Amp gastric, 3rd line 06.001 (20+20mg)
METex14., lung, 3rd line 01.005 (20+20mg) Next scan?
METex14., lung, 1st line 08.001 (20+20mg) Reduction in fatigue and improved breathing reported after 8 days. PR after 7 weeks
METex14., lung, 3rd line 01.004 (60mg) Complete clearance of pleural fluid Reduction in chest pain and weight gain
METex14., lung, 1st line 03.005 (60mg) PD
MET Amp., ovarian, 3rd line 04.001 (40+20mg) PD Non-evaluable: early stop due to sepsis : replace
METex14., lung, ? line 05.001 (40+20mg) 60mg daily
METex14., lung, 2nd line 02.003 (40+20mg) Next scan16/10
METex14., lung, 2nd line 06.002 (40+20mg)
METex14.,lung, 2nd line 08.002 (30+30mg) 13% reduction (RECIST)
METex14.,lung, 2nd line 03.006 (30+30mg) 1st scan?
METex14., lung, 4th line 03.003 (80mg) Non evaluable; protocol violation one kidney : replace
06.003 (40+40mg)
08.002 (40+40mg)
08.002 40+40mg)
80mg daily
Data 15th Oct 2024
>12 weeks Stable Disease (SD) = Disease Control in NSCLC
5 mg once a day
10 mg once a day
20 mg once a day
40 mg once a day
60 mg once a day
80 mg once a day
40 +20 mg BID
20 +20 mg BID
30 +30 mg BID
Drug free interval
Partial Response (PR)
Stable Disease (SD)
Progressive Disease (PD)

- 18 patients dosed
- 9 dose once daily
- 9 dose twice daily (4hrs apart)
- 2 non-evaluable

- Three 1st line patients enrolled
 - 1 PR + 1 SD >10months
 - 100% DCR

Key eligibility criteria

- Proven MET activating mutations or MET amplification (≥ 10 copies) at diagnosis, recent confirmation of MET positivity (tissue or blood) not required at entry
- No longer eligible for approved, available standard therapies
- Eastern Cooperative Oncology Group (ECOG) performance status of 0 or 1

Eligible patients received DO-2 once or twice (4hr interval) daily (QD/BID) orally, under fasting conditions.

- Primary endpoint: safety and tolerability.
- Secondary endpoints: pharmacokinetics and anti-tumor activity.
- Dose-escalation:
 - Simon stage 3 accelerated titration design ("1+1")
 - Starting at 5mg QD, until grade 2 toxicity.
 - Thereafter, a "3+3" design.
- Response assessment was performed every 7-8 weeks.

Patient demographics

Median Age, years (range)	68 (49-77)
Male	13
Female	5
Lung Carcinoma	15
Gastric cancer	1
Kidney Carcinoma	1
Ovarian Carcinoma	1
Stage III	1
Stage IIIB	3
Stage IV	10
Stage IVa	2
Stage IVb	2
MET exon 14 skipping mutation	12
MET amplification positive >10 fold	4
MET activating mutation positive (other than exon 14)	1
Other MET mutation	1
No previous anti-cancer treatment (DO-2 First line)	3
Previously treated with systemic treatment	15
Patients with Measurable Lesions	14
Patients with Non-Measurable Lesions only	3
Unknown	1

Safety of DO-2

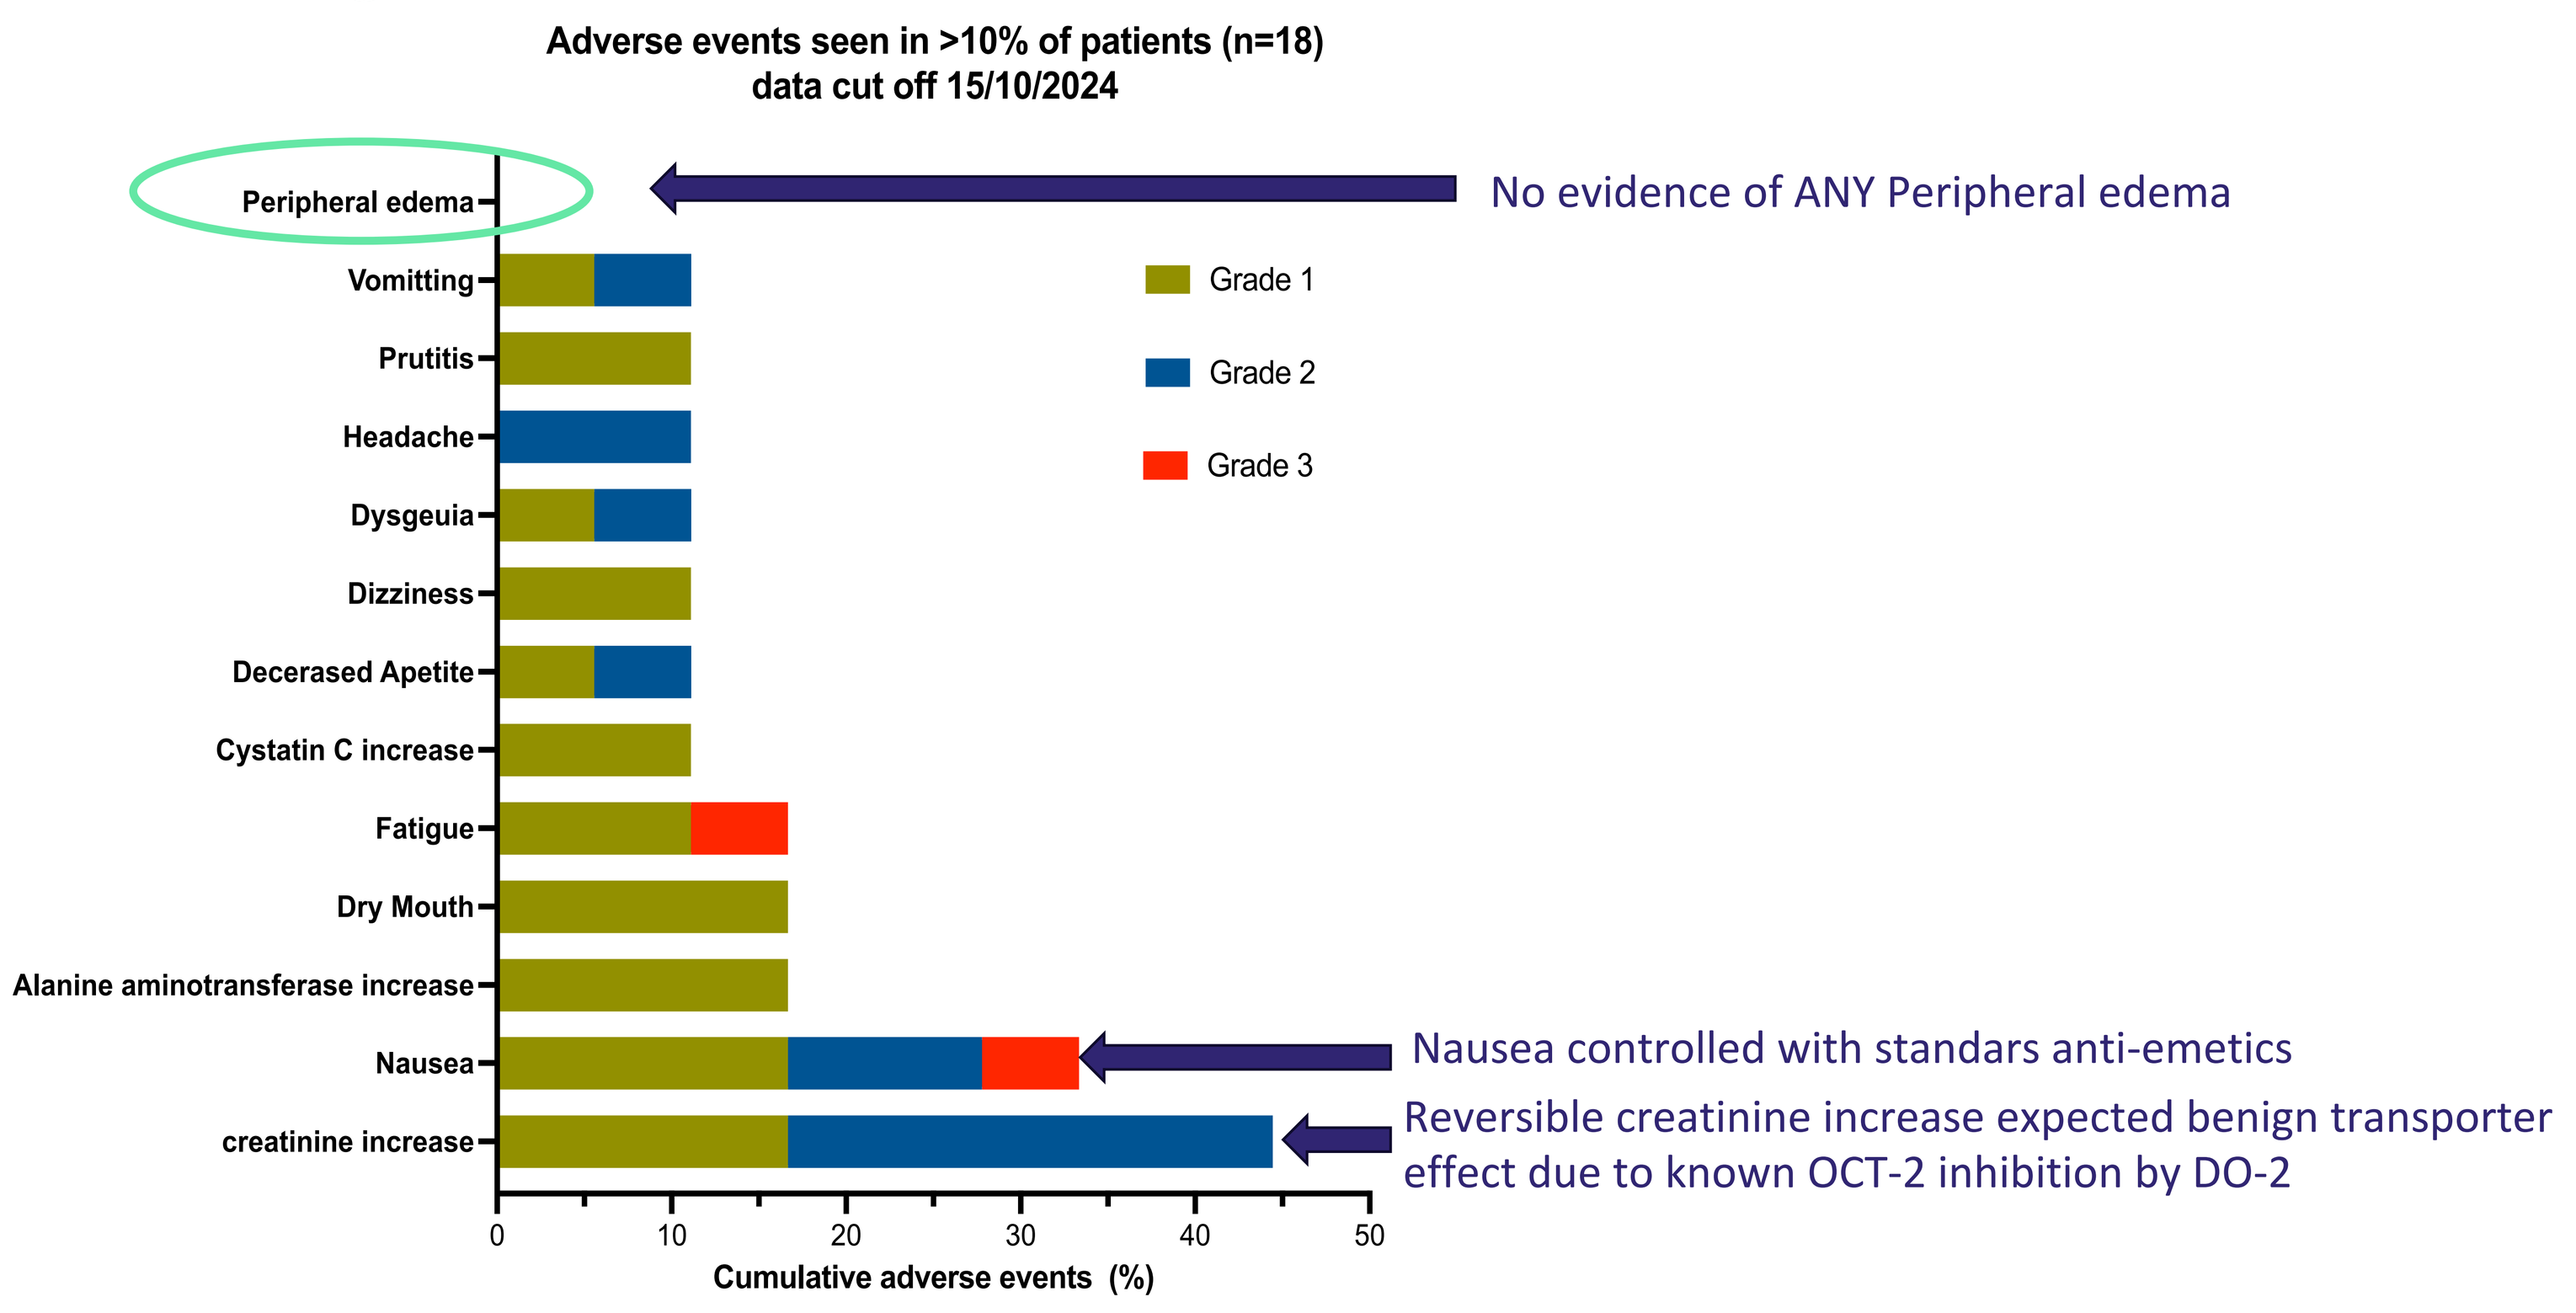
Adverse events seen in >10% of patients (n=18)
data cut off 15/10/2024
Peripheral edema
Vomitting
Prutitis
Headache
Dysgeuia
Dizziness
Decerased Apetite
Cystatin C increase
Fatigue
Dry Mouth
Alanine aminotransferase increase
Nausea
creatinine increase
Grade 1
Grade 2
Grade 3
No evidence of ANY Peripheral edema
Nausea controlled with standars anti-emetics
Reversible creatinine increase expected benign transporter effect due to known OCT-2 inhibition by DO-2
0
10
20
30
40
50
Cumulative adverse events (%)

- Reversible creatinine increase seen in 8/18 patients
- No 'on target' peripheral edema seen
- No liver enzyme elevations seen

Conclusions

- DO-2 is well tolerated at doses up to 60mg QD and 40+20mg BID (4h interval) with no evidence of peripheral edema.
- Reversible creatinine increases (grade 1 and 2) seen in 8/18 patients.
- Partial response (↓39%) seen in 1 patient at 20+20mg and further encouraging signs of clinical benefit, including prolonged (>9 months) disease control observed
- BID dosing (30+30 and 40+40mg) is currently being evaluated.
- Expansion cohorts in patients confirmed to have fresh MET mutation data is foreseen

Disclosures:
The first author has no disclosures of interest.

Study sponsored by DeuterOncology NV
www.deuteroncology.com

Correspondence:
BJ Sikkema, MD
Erasmus MC Cancer Institute Rotterdam
b.sikkema@erasmusmc.nl

Timothy Perera PhD
DeuterOncology SA
tperera@deuteroncology.com

Request a copy